STABILITY INVESTIGATION OF TAMARIND SEED POLYSACCHARIDE UNDER STRESS CONDITIONS

Sabrina Bertini[a], Giorgio Eisele[a], Claudia Foletti[a] , Marco Sansò[b], Giangiacomo Torri[a]
a Ronzoni Institute, Milan, Italy, b Farmigea, Pisa, Italy

INTRODUCTION

Tamarind seed polysaccharide (TSP) xyloglucan is stored in the seed of *Tamarindus Indica*. The primary structure of TSP consists of a (1→4)-b-D-glucan spine with (1→6)-a-branched xylose, which is partially substituted by (1→2)-b-galacto-xylose.

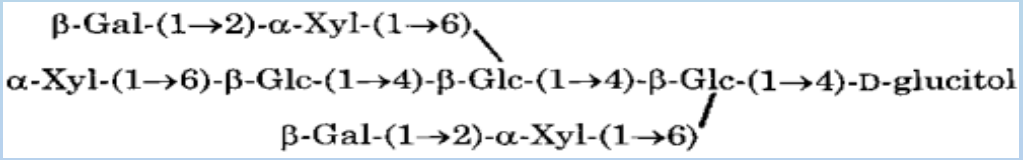

It possesses properties like high viscosity, broad pH tolerance and adhesivity, so this led to its application as stabilizer, thickener, gelling agent and binder in food and pharmaceutical industries [1, 2]. In addition to these, it has characteristics of non-carcinogenicity, mucoadhesivity, biocompatibility and high drug holding capacity [3].Since TSP is an important excipient, the present study was undertaken to elucidate the stability of the polysaccharide.

We solubilized TSP in different conditions to verify the stability and characterized potential products of degradation at:

- high temperature
- basic and acidic pH
- high pressure
- presence of hydroxyl radicals

To determine the structure of TSP before and after treatments, different technique have been applied. High Field Nuclear Magnetic Spectroscopy has been performed to investigate the chemical modifications of primary structure of TSP while High Performance Size Exclusion Chromatography combined with four on-line detectors, right and low angle light scattering, refractometer and viscometer (HP-SEC-TDA) has been done for the evaluation of Mn (number-average mean molecular weight), Mw (weight-average mean molecular weight) and Mw/Mn (polydispersity), but also for evaluating hydrodynamic radius (Rh) [4]. Moreover, the hydrodynamic radius of TSP in solution was obtained directly from Dynamic Light Scattering measurements and the results have been compared with HP-SEC-TDA 's one.

RESULTS

[1]H-NMR analysis:

To characterize primary structure of TSP and investigate its chemical modifications, NMR analysis was performed and ^{1}H-spectra of solution were recorded. By comparing NMR spectra of samples solubilized in different conditions, we observed new peaks at 8.5-8.0 ppm and 3-0 ppm (*).

These new signals could be attributed to organic acid and aliphatic chains formed due to the degradation of sugar units. This phenomena appears more evident with the increasing temperature, time of solubilization and also pH. Under acidic condition, we noticed the presence of signals at about 5-5.5ppm that could be attributed to oligosaccharides units(§).

In high pressure treatment, these signals were absent (there were no change in the spectra); this suggests the TSP's stability in these stress conditions.

When TSP undergone radical reactions we observed peaks only at 8.5-8.0 ppm due to the oxidative degradation.

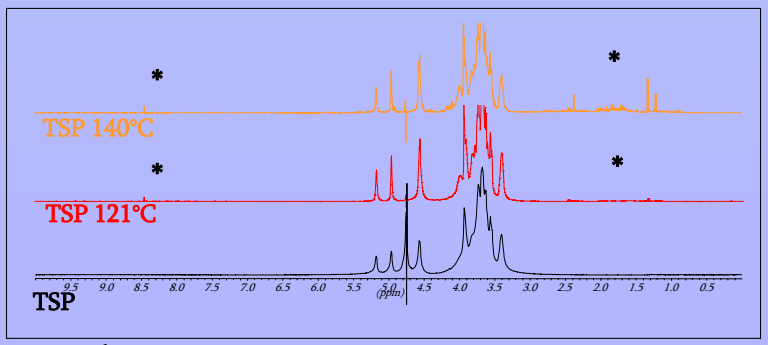
TSP 140°C
TSP 121°C
TSP

Figure 1: ^{1}H-NMR spectra of TSP after treatment at high temperature

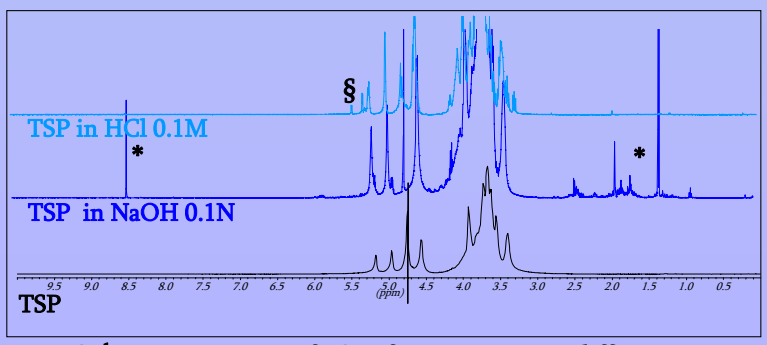
§
TSP in HCl 0.1M
TSP in NaOH 0.1N
TSP

Figure 2: ^{1}H-NMR spectra of TSP after treatment at different pH

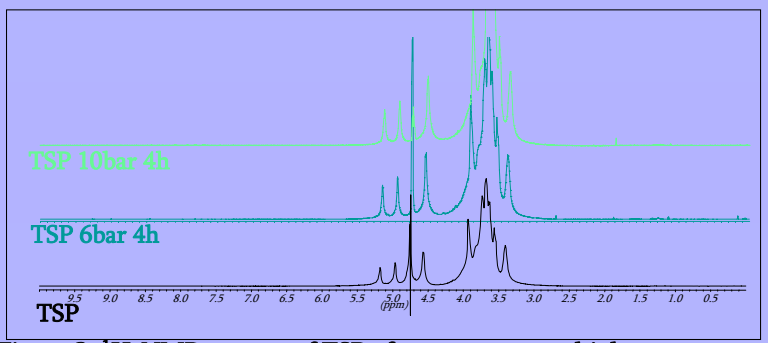
TSP 10bar 4h
TSP 6bar 4h
TSP

Figure 3: ^{1}H-NMR spectra of TSP after treatment at high pressure

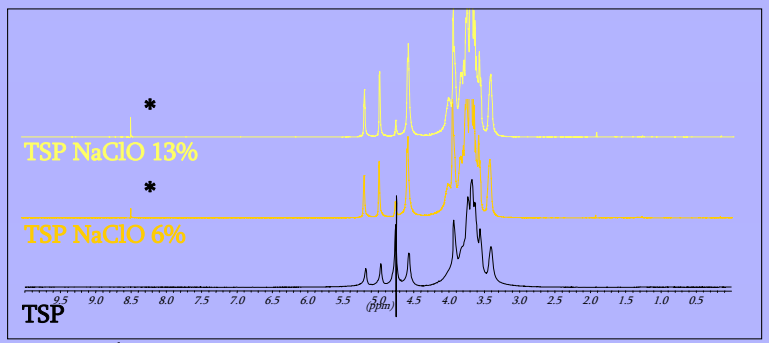
TSP NaClO 13%
TSP NaClO 6%
TSP

Figure 4: ^{1}H-NMR spectra of TSP after treatment in presence of hydroxyl radicals

HP-SEC-TDA analysis:

Sample	Mw (Da)	Mw/Mn	[η] (dl/g)	Rh (nm)
TSP	627.500	1.7	6.3	37.51
TSP 121°C 8h	67.100	1.68	1.0	10.4
TSP 140°C 8h	28.800	1.43	0.5	5.6

Table 1: results from HP-SEC-TDA analysis of TSP after treatment in high temperature

Sample	Mw (Da)	Mw/Mn	[η] (dl/g)	Rh (nm)
TSP in NaOH 0.1N 8h	249.700	2.91	1.5	21.73
TSP in HCl 0.1M 8h	18.300	2.00	0.5	4.69

Table 2: results from HP-SEC-TDA analysis of TSP after treatment at different pH

Sample	Mw (Da)	Mw/Mn	[η] (dl/g)	Rh (nm)
TSP 6bar	717.700	1.66	6.7	40.48
TSP 10bar	734.500	1.58	6.8	41.16

Table 3: results from HP-SEC-TDA analysis of TSP after treatment at high pressure

Sample	Mw (Da)	Mw/Mn	[η] (dl/g)	Rh (nm)
TSP NaClO 6%	69.300	1.93	0.96	9.45
TSP NaClO 13%	29.400	1.90	0.49	5.78

Table 4: results from HP-SEC-TDA analysis of TSP after treatment in high temperature

1) Both molecular weight (Mw) and intrinsic viscosity [η] have a progressive decrease in relation with the increase of reaction's temperature. In the table1 values obtained from elaboration performed with software OminSEC 4.2 are shown.

2) Both Mw and IV decreased in condition of basic pH (NaOH 0.01N and 0.1N). Instead in the case of acid pH we noticed a decrease of these values only in extreme condition (HCl 0.1M), infect TSP structure does not change significantly performing solubilization in TFA 0.2M. In table 2 we reporting data of TSP after treatment in extreme basic and acid pH conditions.

3) Structure's parameters of TSP do not change after treatment at high pressure (6 and 10bar). This support data obtained from NMR analysis and demonstrate the polysaccharide's stability in these stress conditions.

4) Reaction in presence of hydroxyl radicals produces a strong degradation of TSP.

Dynamic light scattering analysis:

Dynamic Light Scattering system measures light diffusion deviated by molecules throw light scattering at 90°. These data was then correlated by an algorithm at hydrodynamic radius of every species present into the solution.

The molecular weight with the equation $Rh = K*M^a$; for TSP we had determined *a* and *K* parameters by HP-SEC-TDA analysis. Data that we obtained demonstrate a substantial agreement between the two method of analysis, both for Rh and Mw. In figure 5 we reported as example data from treatment at 121°C.

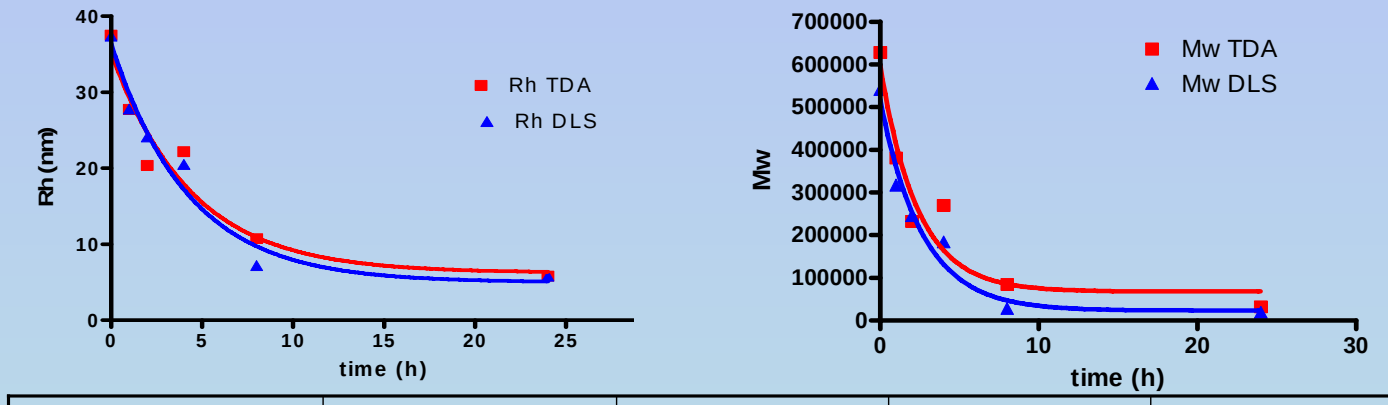
Rh (nm)
time (h)
Rh TDA
Rh DLS
Mw
time (h)
Mw TDA
Mw DLS

Time (h) 121°C	Rh (nm) TDA	Rh (nm) DLS	Mw (Da) TDA	Mw (Da) DLS
0	**37,5**	**37,4**	**627.500**	**541.000**
1	27,7	27,8	380.200	317.000
2	20,4	24,1	231.600	246.000
4	22,2	20,5	269.000	184.000
8	10,8	7,2	84.000	28.000
24	5,8	5,8	31.500	19.000

Figure 5: Comparison of data from TDA and DLS

CONCLUSIONS

From this work we can conclude:

1-TSP is stable at high pressure.

2-TSP undergoes degradation to low molecular weight fractions, oligosaccharide and monosaccharide only at extreme conditions of pH and temperature

3-Radicals also cause significant degradation of TSP.

REFERENCES

1) Rao PS, Ghosh TP, Krishna S; J Sci Ind Research1946; 4: 705
2) Sumathi S, Ray AR; J Pharm Pharm Sci. 2002 Jan-Apr;5(1):12-8
3) Saettone MF, Burgalassi S, Giannaccini B et al.; PCT Int Appl WO 97 28, 787, 1997
4) Liu XM, Gao W, Maziarz EP et al.; J Chromatogr A. 2006 Feb 3;1104(1-2):145-53